P.7.d.014

Guanfacine extended release: daytime sleepiness outcomes from a phase 3 clinical study in adolescents with attention-deficit/ hyperactivity disorder

Andrew Cutler,[1] Linda Harper,[2] Joel L Young,[3] Ben Adeyi,[4] Bryan Dirks,[4] Timothy Wilens[5]

[1]Florida Clinical Research Center, Bradenton, FL, USA; [2]CNS Healthcare, Orlando, FL, USA; [3]Rochester Center for Behavioral Medicine, Rochester Hills, MI, USA; [4]Shire, Wayne, PA, USA; [5]Massachusetts General Hospital, Boston, MA, USA

Scan code to receive PDF file of the poster
or visit: www.shirecongressposters.com/xxxxxx

INTRODUCTION

- Guanfacine extended-release (GXR), a long-acting, non-stimulant, selective α_{2A}-adrenergic receptor agonist,[1–3] is approved as a monotherapy or adjunctive therapy to stimulants for the treatment of attention-deficit/ hyperactivity disorder (ADHD) in children and adolescents (aged 6–17 years) in the USA and in children (aged 6–12 years) in Canada.
- A phase 3 study (SPD503-312) demonstrated that GXR was more effective than placebo in reducing ADHD Rating Scale IV (ADHD-RS-IV) total scores from baseline to endpoint (primary outcome) in adolescents (aged 13–17 years).[4]
- GXR is associated with sedation-related adverse events.[5]

OBJECTIVE

- To report on Pediatric Daytime Sleepiness Scale (PDSS) scores and sedation-related treatment emergent adverse events (TEAEs) in a phase 3 study of GXR treatment in adolescents with ADHD (SPD503-312).

METHODS

Study design

- SPD503-312 was a 13-week, randomized, double-blind, multicentre, parallel-group, dose-optimized, phase 3 study comparing GXR with placebo (Figure 1).
- This study enrolled adolescents (aged 13–17 years) with a primary diagnosis of ADHD based on the Diagnostic and Statistical Manual of Mental Disorders, Fourth Edition, Text Revision criteria.
 - Eligible participants had a baseline ADHD-RS-IV total score of at least 32 and a baseline Clinical Global Impressions-Severity (CGI-S) score of at least 4.
 - Participants were enrolled at 48 sites in the USA; four additional sites conducted screening but did not enrol patients.
- Individuals were randomized 1:1 to receive once-daily GXR or placebo for a 7-week dose-optimization period followed by a 6-week dose-maintenance period.
 - During the optimization period, the GXR dose was adjusted until an 'acceptable' response was achieved, defined as at least a 30% reduction from baseline in ADHD-RS-IV total score, and a Clinical Global Impressions-Improvement (CGI-I) score of 1 or 2, with tolerable side effects.
 - The maximum GXR dose was in the range of 4–7 mg/day (up to 0.12 mg/kg/day), depending on patient weight and tolerance of the drug.

Analysis of sedation-related outcomes

- In SPD503-312, self-reported daytime sleepiness was a secondary outcome and was assessed using the PDSS questionnaire at baseline and throughout the treatment period.
 - The questionnaire consists of eight questions that are scored using a Likert scale from 0 (never) to 4 (always/very often).
- TEAEs relating to sedation (i.e. somnolence, sedation and hypersomnia, were considered of clinical importance.

Figure 1. Study design.

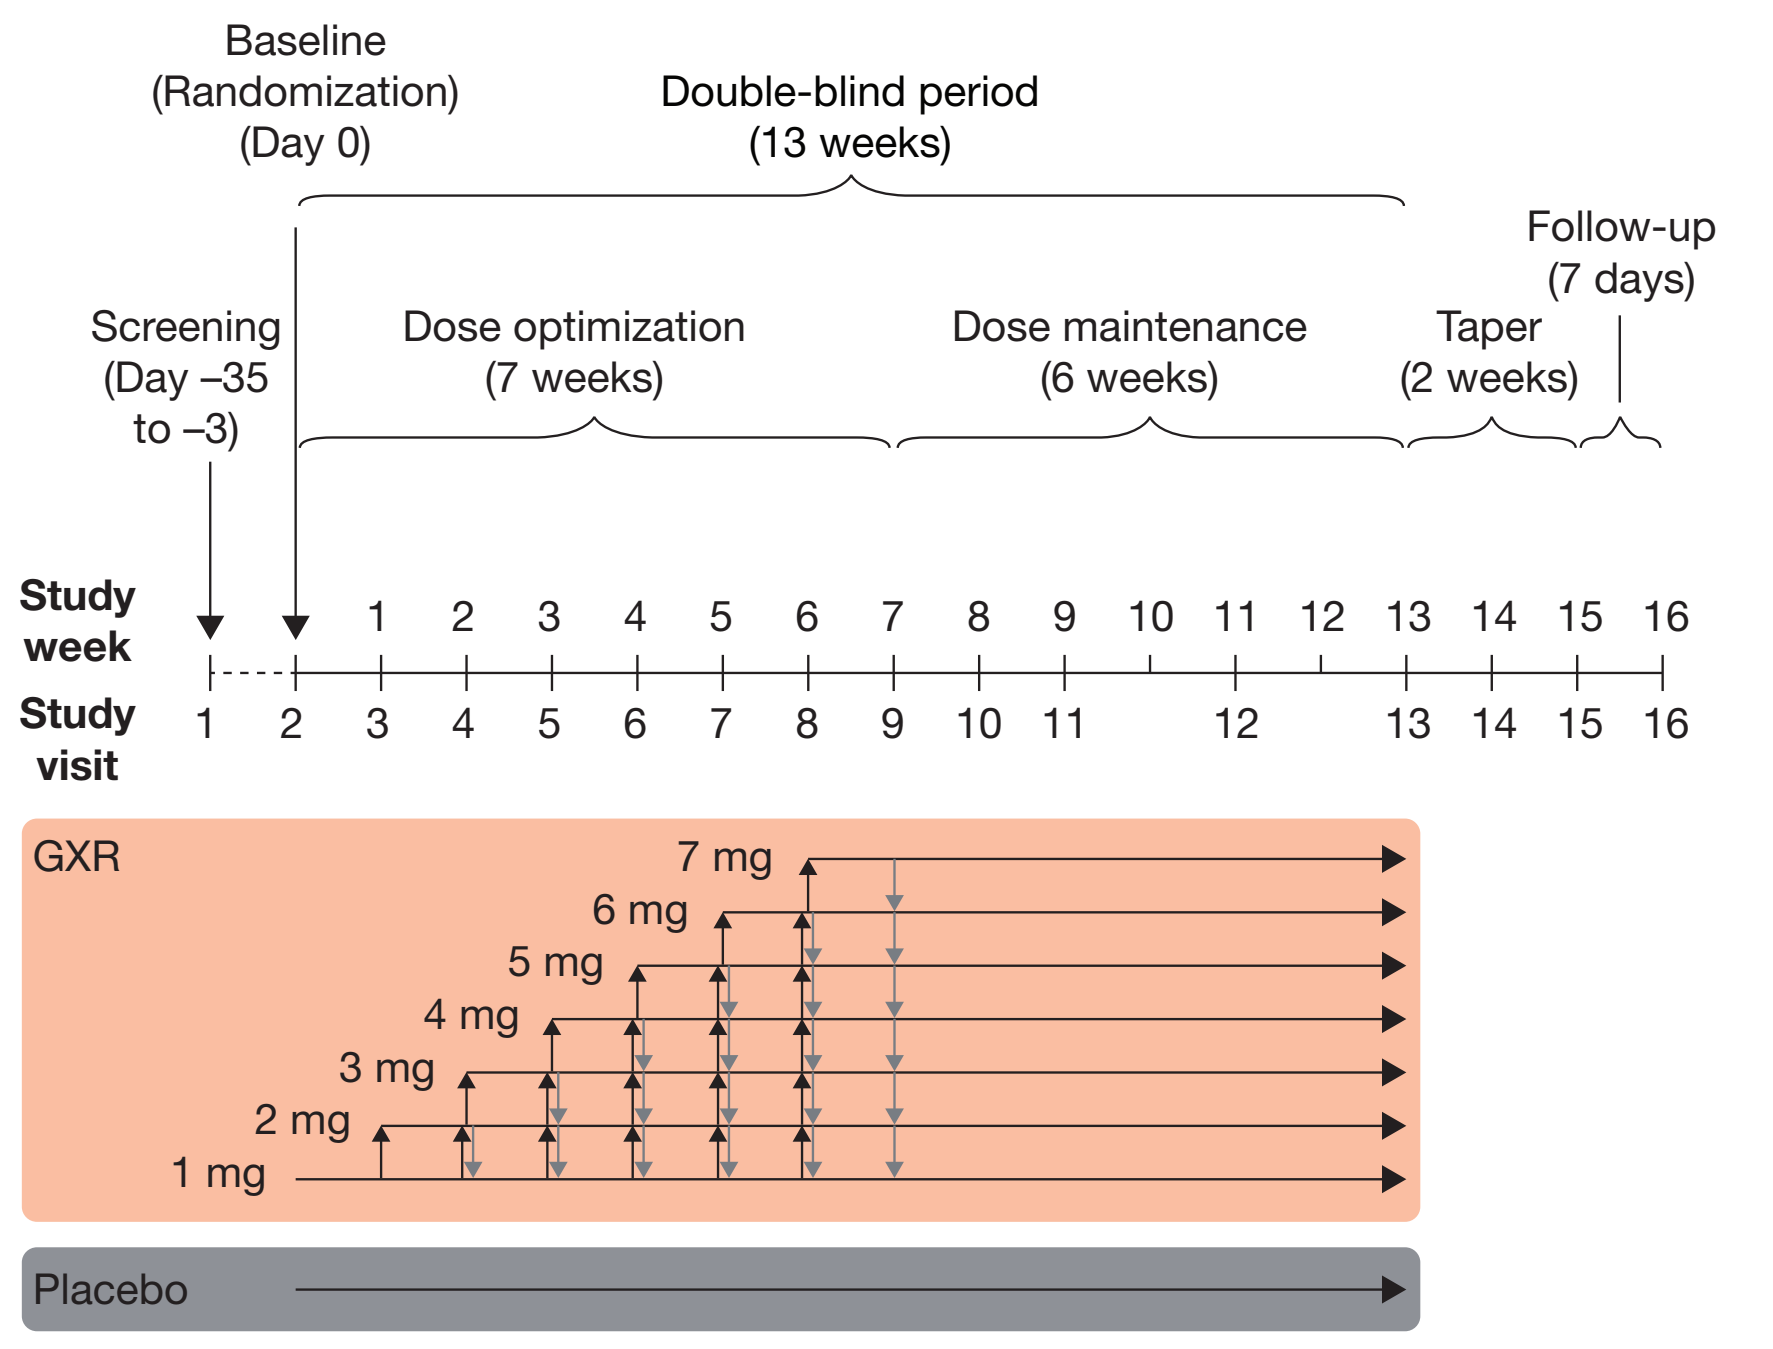

Baseline
(Randomization)
(Day 0)
Double-blind period
(13 weeks)
Follow-up
(7 days)
Screening
(Day –35
to –3)
Dose optimization
(7 weeks)
Dose maintenance
(6 weeks)
Taper
(2 weeks)
Study week
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16
Study visit
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16
GXR
7 mg
6 mg
5 mg
4 mg
3 mg
2 mg
1 mg
Placebo

Individuals assigned to GXR could receive up to 0.12 mg/kg/day, with dose limits of 4 mg, 5 mg, 6 mg and 7 mg for weight groups 34.0–41.4, 41.5–49.4, 49.5–58.4 and 58.5–91.0 kg, respectively. All individuals initially received GXR 1 mg/day or placebo, starting the morning after the baseline visit (visit 2). Dose optimization was carried out in single-step titrations as shown above; doses could be lowered once to the previous dose level, but could not be adjusted after visit 9. Endpoint was the last valid assessment obtained after baseline while on investigational product and before the first dose-taper.
GXR, guanfacine extended release.

Table 1. Key demographics and baseline disease characteristics (safety population, N = 312)

Characteristic	GXR (n = 157)	Placebo (n = 155)
Age, years, mean (SD)	14.5 (1.35)	14.6 (1.44)
Sex, male, n (%)	103 (65.6)	99 (63.9)
Race, white, n (%)	113 (72.0)	114 (73.5)
BMI, kg/m², mean (SD)	22.00 (3.343)	21.69 (3.239)
Baseline ADHD-RS-IV total score, mean (SD)	39.9 (5.57)	40.0 (6.11)

The safety population comprised all individuals who were randomized and took at least one dose of study drug.
ADHD-RS-IV, ADHD Rating Scale IV; BMI, body mass index; GXR, guanfacine extended release; SD, standard deviation.

Figure 2. Change from baseline in mean (SD) PDSS total score at each study visit (safety population, N = 312).

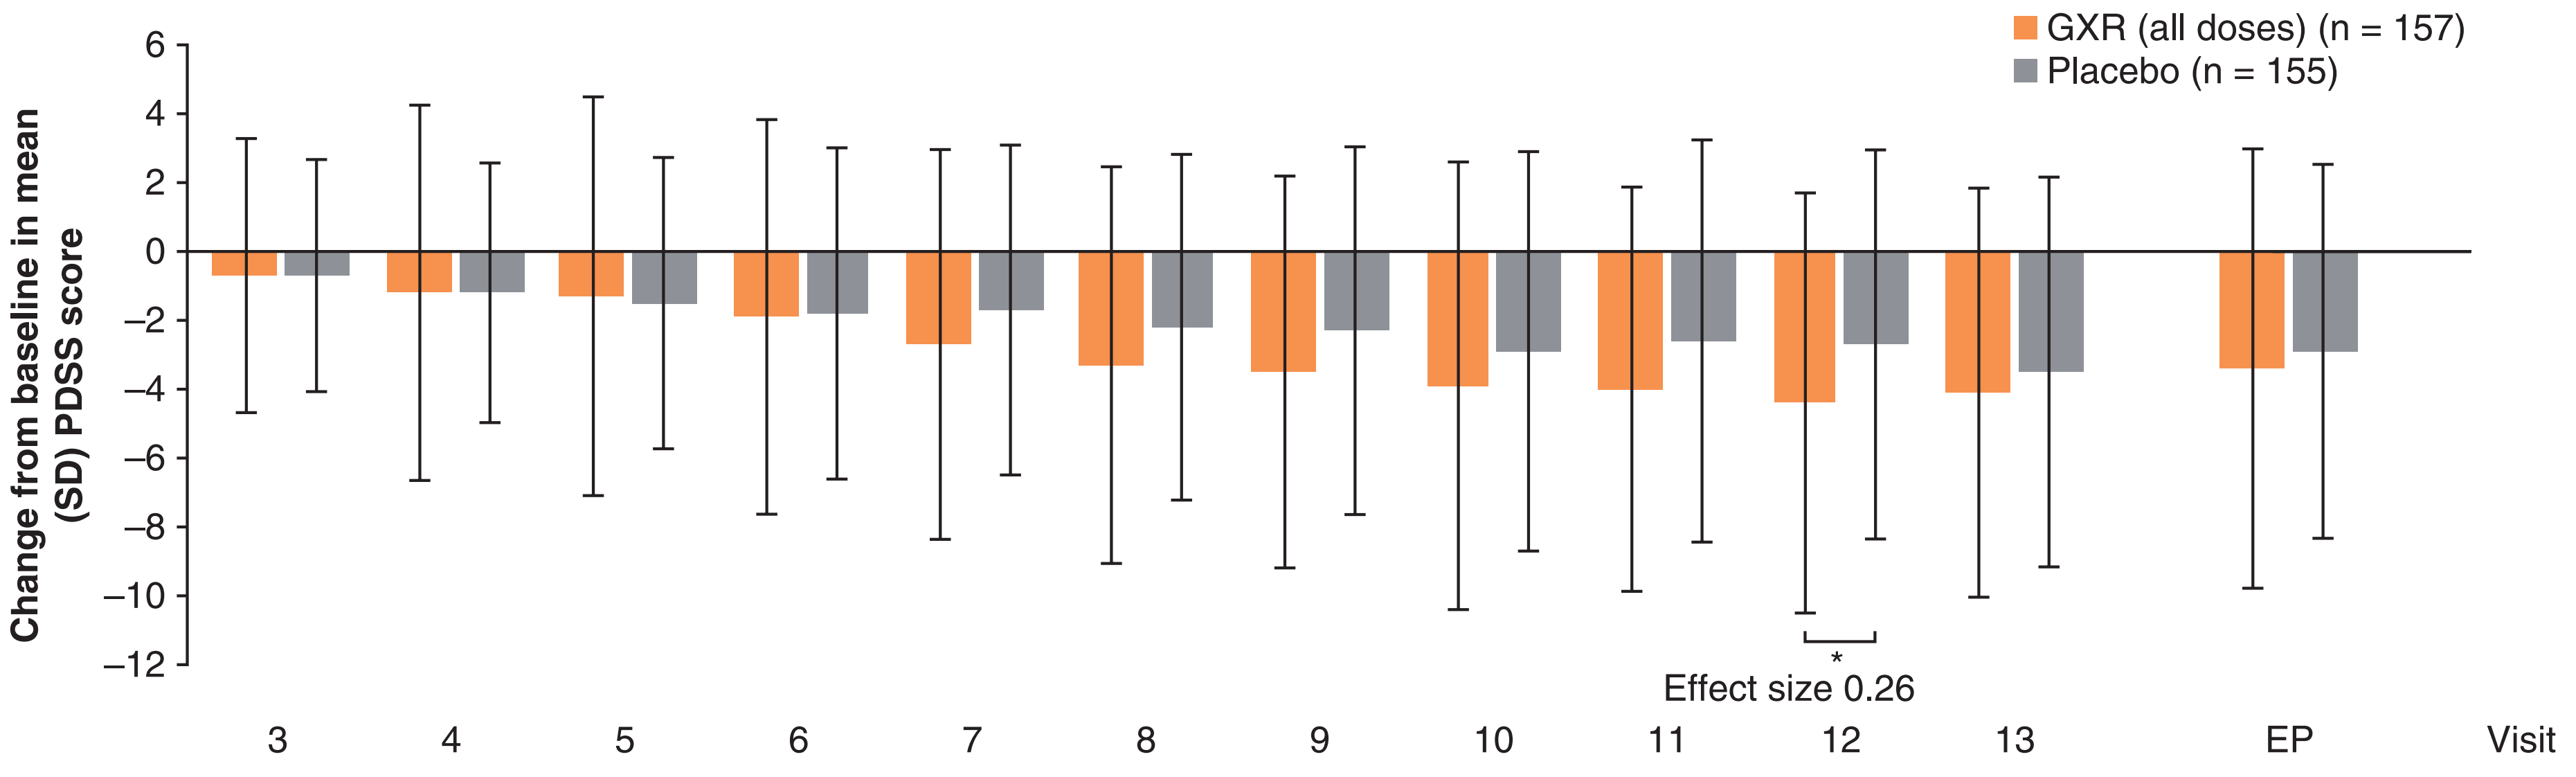

GXR (all doses) (n = 157)
Placebo (n = 155)
Change from baseline in mean (SD) PDSS score
6
4
2
0
–2
–4
–6
–8
–10
–12
Effect size 0.26
*
3 4 5 6 7 8 9 10 11 12 13 EP
Visit

*$p < 0.05$, based on the difference between GXR and placebo in LS mean change from baseline.
Endpoint is the last valid assessment obtained after baseline while on investigational product and before first dose-tapering step.
EP, endpoint; GXR, guanfacine extended release; LS, least-squares; PDSS, Pediatric Daytime Sleepiness Scale; SD, standard deviation.

Figure 3. Incidence of sedation-related adverse events (somnolence, sedation and hypersomnia) in the safety population (N = 312).

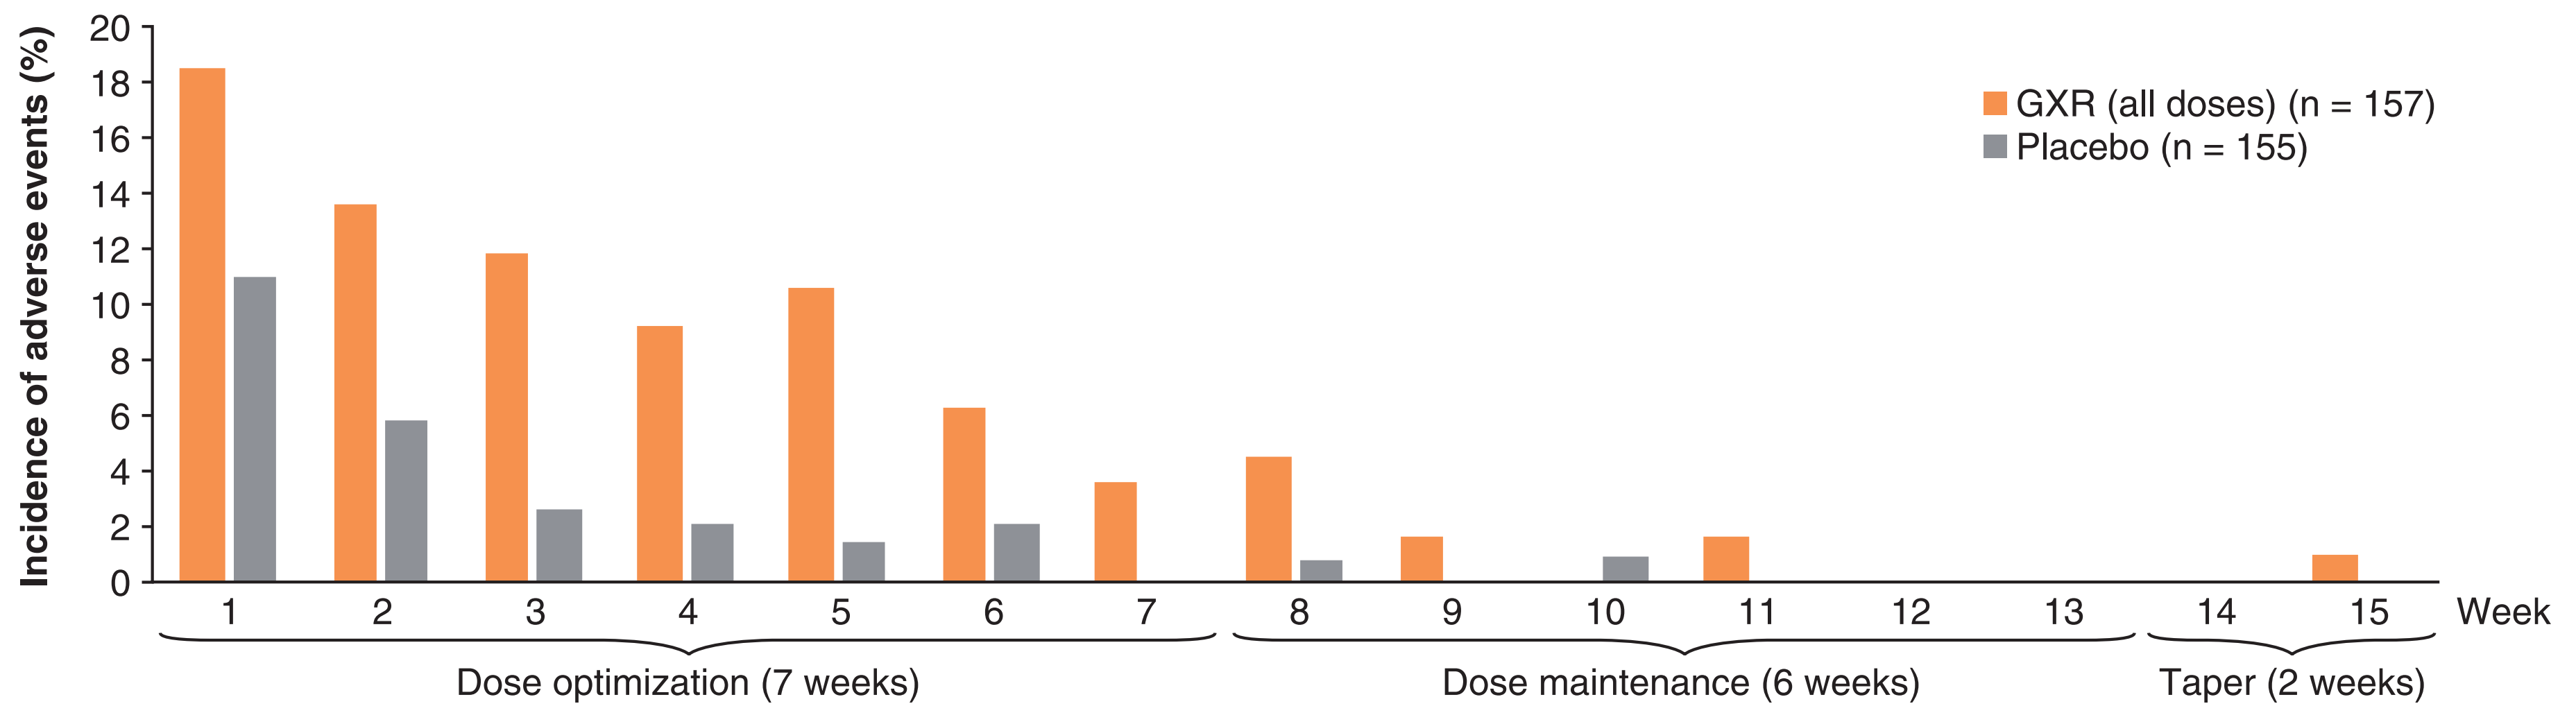

GXR (all doses) (n = 157)
Placebo (n = 155)
Incidence of adverse events (%)
20
18
16
14
12
10
8
6
4
2
0
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15
Week
Dose optimization (7 weeks)
Dose maintenance (6 weeks)
Taper (2 weeks)

GXR, guanfacine extended release.

RESULTS

Patient demographics and baseline characteristics

- In total, 314 individuals were randomized to GXR (n = 157) or placebo (n = 157). Of these, 312 were included in both the full analysis set (FAS) and the safety population (GXR, n = 157; placebo, n = 155), and 227 completed to visit 13 (GXR, n = 117; placebo, n = 110).
- Patient demographics and baseline characteristics in the GXR and placebo groups were similar (Table 1).
- The mean (standard deviation [SD]) optimal and weight-adjusted doses of GXR at visit 9 were 4.3 (1.50) mg and 0.073 (0.0248) mg/kg, respectively.

Pediatric Daytime Sleepiness Scale total score

- The changes from baseline in mean PDSS total score for GXR and placebo groups at each study visit are shown in Figure 2.
 - A negative change in PDSS score indicates a reduction in self-reported daytime sleepiness compared with baseline.
- The difference (GXR minus placebo) in least-squares (LS) mean change from baseline was statistically significant ($p = 0.038$; effect size, 0.26) at visit 12 only, not at study endpoint.

Table 2. Frequently occurring (≥ 5%) TEAEs (safety population, N = 312

Preferred term, n (%)	GXR (n = 157)	Placebo (n = 155)
Any TEAE	147 (93.6)	120 (77.4)
Serious	4 (2.5)	2 (1.3)
Related to study drug	125 (79.6)	80 (51.6)
Leading to discontinuation	9 (5.7)	3 (1.9)
Leading to dose reduction	21 (13.4)	8 (5.2)
Leading to death	0 (0.0)	0 (0.0)
TEAEs reported by ≥ 5% of participants in any treatment group		
Somnolence	69 (43.9)	33 (21.3)
Headache	42 (26.8)	28 (18.1)
Fatigue	35 (22.3)	19 (12.3)
Dizziness	25 (15.9)	16 (10.3)
Decreased appetite	23 (14.6)	21 (13.5)
Nausea	19 (12.1)	21 (13.5)
Nasopharyngitis	18 (11.5)	9 (5.8)
Sedation	18 (11.5)	3 (1.9)
Increased appetite	14 (8.9)	13 (8.4)
Insomnia	14 (8.9)	6 (3.9)
Upper respiratory tract infection	14 (8.9)	12 (7.7)
Diarrhoea	12 (7.6)	13 (8.4)
Dry mouth	12 (7.6)	0 (0.0)
Irritability	11 (7.0)	6 (3.9)
Abdominal pain upper	10 (6.4)	7 (4.5)
Abdominal pain	9 (5.7)	6 (3.9)
Vomiting	9 (5.7)	10 (6.5)
Dizziness postural	8 (5.1)	3 (1.9)
Cough	3 (1.9)	8 (5.2)

Treatment-emergent adverse events

- TEAEs and TEAEs reported by at least 5% of study participants during the study period are summarized in Table 2.
- Most TEAEs were mild or moderate. Serious TEAEs were reported by 4 individuals (2.5%) receiving GXR and 2 (1.3%) receiving placebo.
- No deaths were reported.
- TEAEs leading to discontinuation were reported by 9 individuals (5.7%) receiving GXR and 3 (1.9%) receiving placebo.
- The TEAEs reported by at least 5% of study participants were consistent with the known effects of GXR.

Sedation-related adverse events

- At least one sedative event (somnolence, sedation or hypersomnia) was reported in 54.1% (85/157) of those receiving GXR and 22.6% (35/155) of those receiving placebo.
- In the safety population, the majority of sedative events were mild or moderate, and most resolved before the end of the dose-maintenance period (Figure 3).
- One case of somnolence in the GXR group led to discontinuation from the study.

CONCLUSIONS

- In this study, GXR was generally well tolerated and the safety profile was similar to that seen in other studies of GXR.
 - Despite the higher incidence of sedation-related adverse events in the GXR group (at doses up to 7 mg/day) than in the placebo group, there was no difference in the PDSS total score between the two groups throughout the treatment period, with the exception of visit 12.
- All sedation-related adverse events were transient and resolved during the study period.

REFERENCES

1. Biederman J *et al. Pediatrics* 2008;121:e73–84.
2. Sallee FR *et al. J Am Acad Child Adolesc Psychiatry* 2009;48:155–65.
3. Michelson D *et al. Am J Psychiatry* 2002;159:1896–901.
4. Wilens TE *et al.* Presented at the 3rd Eunethydis International Conference on ADHD, 21–24 May 2014, Istanbul, Turkey.
5. Intuniv® prescribing information. Available from: http://pi.shirecontent.com/PI/PDFs/Intuniv_USA_ENG.pdf (Accessed 19 June 2015).

Disclosures

B Dirks is an employee of Shire Development LLC and holds stock or stock options. B Adeyi is a former employee of Shire. The following authors have received compensation for serving as consultants or speakers for, or they or the institutions they work for have received research support or royalties from, the companies or organizations indicated: A Cutler (AbbVie [Abbott], Akili Interactive, Arbor Pharmaceuticals, AstraZeneca, Euthymics, Janssen, Johnson & Johnson PRD, Lilly, Neos Therapeutics, Neurovance, Novartis, Noven, Otsuka, Pfizer [NextWave], Purdue, Rhodes Pharmaceuticals, Shionogi, Shire, Sunovion, Supernus, Targacept and Theravance); L Harper (Forest and Takeda); T Wilens (US National Institutes of Health [NIDA], Euthymics/ Neurovance, Ironshore, Sunovion, Theravance and TRIS); JL Young (Eli Lilly, Forest, Otsuka, Pfizer, Shire, Sunovion Takeda/Lundbeck and Teva).

Writing and editing assistance was provided by Oxford PharmaGenesis, UK and funded by Shire International GmbH.

The authors thank the patients, parents/guardians and investigators who participated in the study.

Mobile and environmentally friendly e-posters

3 ways to access the poster

Send an SMS
Text 'Poster xxxxxx' to +44 786 003 3046

Access online
http://www.shirecongressposters.com/xxxxxx

Scan the QR code

No personal information is stored – standard charges may apply

Study funded by Shire Development LLC.

Presented at the 28th European College of Neuropsychopharmacology Congress, 29 August–1 September 2015, Amsterdam, Netherlands

SPIV045-Poster 06 New template blue and blue.indd 1

03/08/2015 17:47